Population Pharmacokinetic Model for Human Growth Hormone in Adult Patients in Chronic Dialysis vs. Healthy Subjects

T Klitgaard[1,✉] , JN Nielsen[1], MS Fitsios[1], and M Lange[2]

[1]Novo Nordisk A/S Bagsvaerd, Denmark;[2]Clinical Research, Novo Nordisk Inc., Princeton, US. ✉ tkt@novonordisk.com

Background and Objectives

- Recombinant human Growth Hormone (rhGH) may be beneficial in treating adult patients in chronic dialysis (APCD pts.). Since both the kidneys and the liver are reported to play a role in GH clearance, pharmacokinetics could be significantly influenced by renal function impairment, with possible implications on accumulation and, in turn, efficacy and safety.
- Objective: To develop a population pharmacokinetic (popPK) model for rhGH in APCD pts. and healthy volunteers (HVs), to support the design of future clinical trials.

Trial design and subjects

- **Design:** Open, non-randomized, single-center, parallel-group study over 8-9 days.
- **Subjects:** 11 APCD pts., 10 matched healthy controls.
- **Dose:** 50 µg/kg/day rhGH s.c. for 7 days. ERSD pts. had an extra dose on Day 8 and 4 dialysis sessions over the 9 day period.

Figure 1. Trial Design

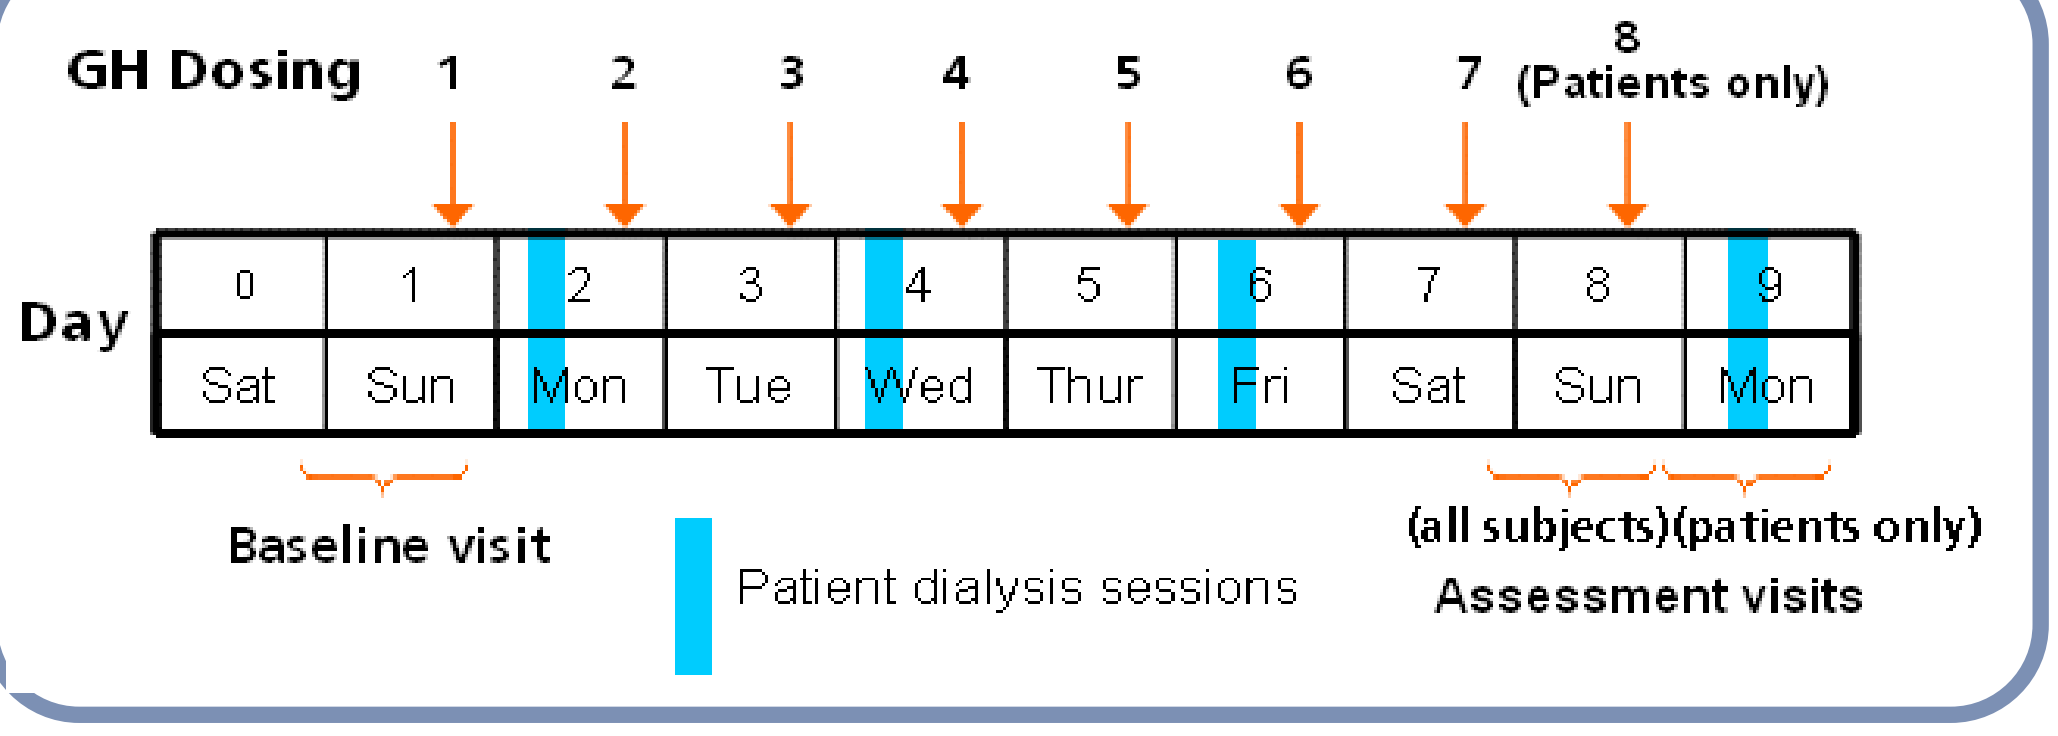
GH Dosing
1
2
3
4
5
6
7
8 (Patients only)
Day
0 1 2 3 4 5 6 7 8 9
Sat Sun Mon Tue Wed Thur Fri Sat Sun Mon
Baseline visit
Patient dialysis sessions
(all subjects)(patients only)
Assessment visits

Methods

- Analysis was performed in NONMEM V. First order conditional (FOCE) estimation method with INTERACTION was used.
- The following structural models were evaluated:
 - One- and two- compartment (CMT) models with first-order (FO) absorption and elimination
 - One CMT models with FO absorption and Michaelis-Menten (MM) type of elimination
 - One CMT models with FO absorption and MM elimination + parallel FO elimination
 - One CMT models with MM absorption and elimination +/- parallel FO elimination
 - All above +/- individual absorption lag time or +/- absorption delay CMT
- Covariates APCD/HV, Gender, Weight, and Dialysis Flow rate (APCD pts. only) were tested on key model parameters.
- Exponential error models used for inter-individual variability. Combined error model used for residual error.
- Model performance was evaluated with a simplified posterior predictive check: 1000 trials were simulated and distributions of mean, geometric mean, and standard deviation of AUC0-24h and Cmax were compared with non-compartmental estimates.
- Population mean profiles with 95% confidence limits were simulated for each of the two groups of subjects.

Figure 2. Final Model

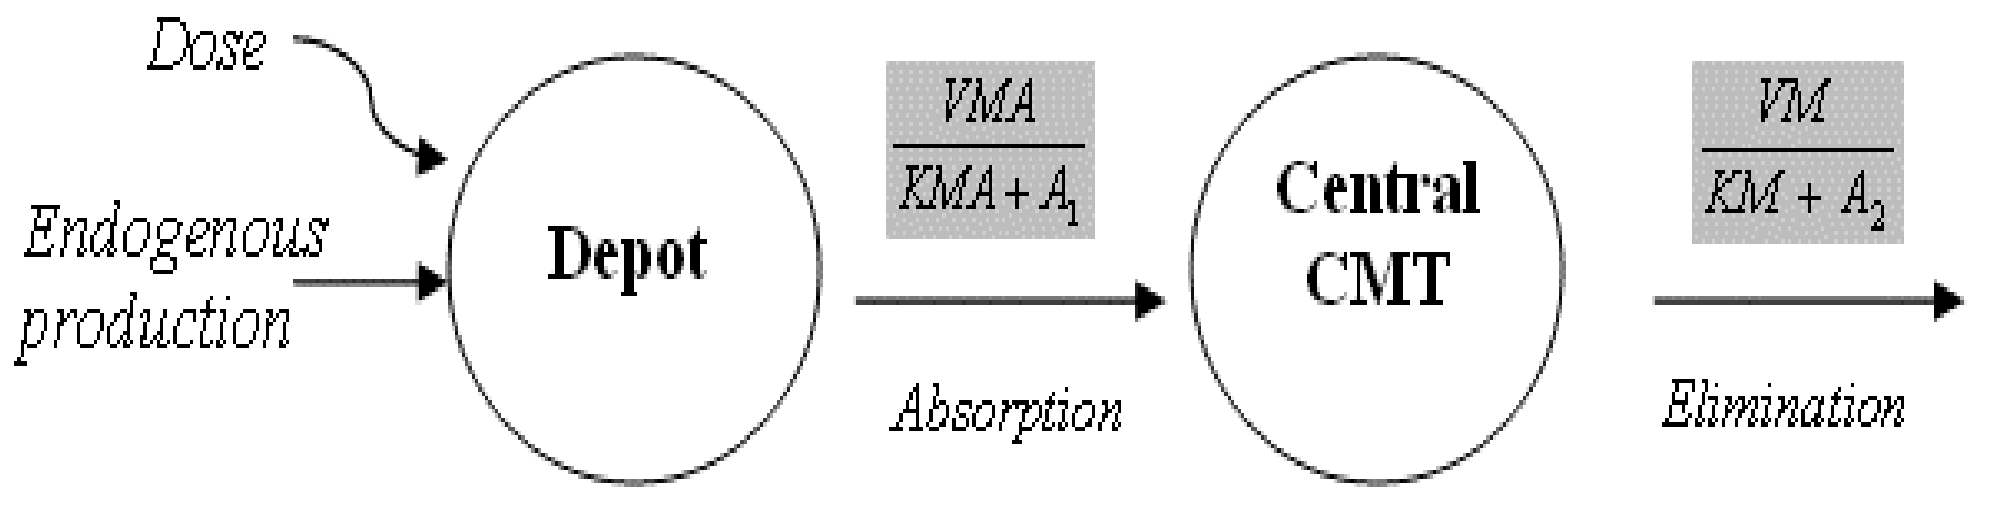
Dose
Endogenous production
Depot
VMA / KMA + A1
Absorption
Central CMT
VM / KM + A2
Elimination

One CMT model with MM absorption and elimination, and an individual baseline level of endogenous hGH. A1: Drug amount in depot, A2: Drug amount in central compartment, VMA, VM: Max absorption, elimination rate, KMA, KM: Amount for half-max absorption, elimination rate

Results

- Final model was a one CMT model with MM-absorption and MM-elimination The latter possibly describes both a FO (renal) and a MM (hepatic) elimination (could not be separated).
- Statistically significant ($p<0.001$) differences in parameters for absorption (KMA) and elimination (VM) for HVs vs. APCD pts.
- A posterior predictive check indicated acceptable performance for simulation purposes, to support future studies.
- Large inter-individual variation, as reflected in simulated mean profiles with 95% confidence limits.

Figure 3. Simplified Posterior predictive check

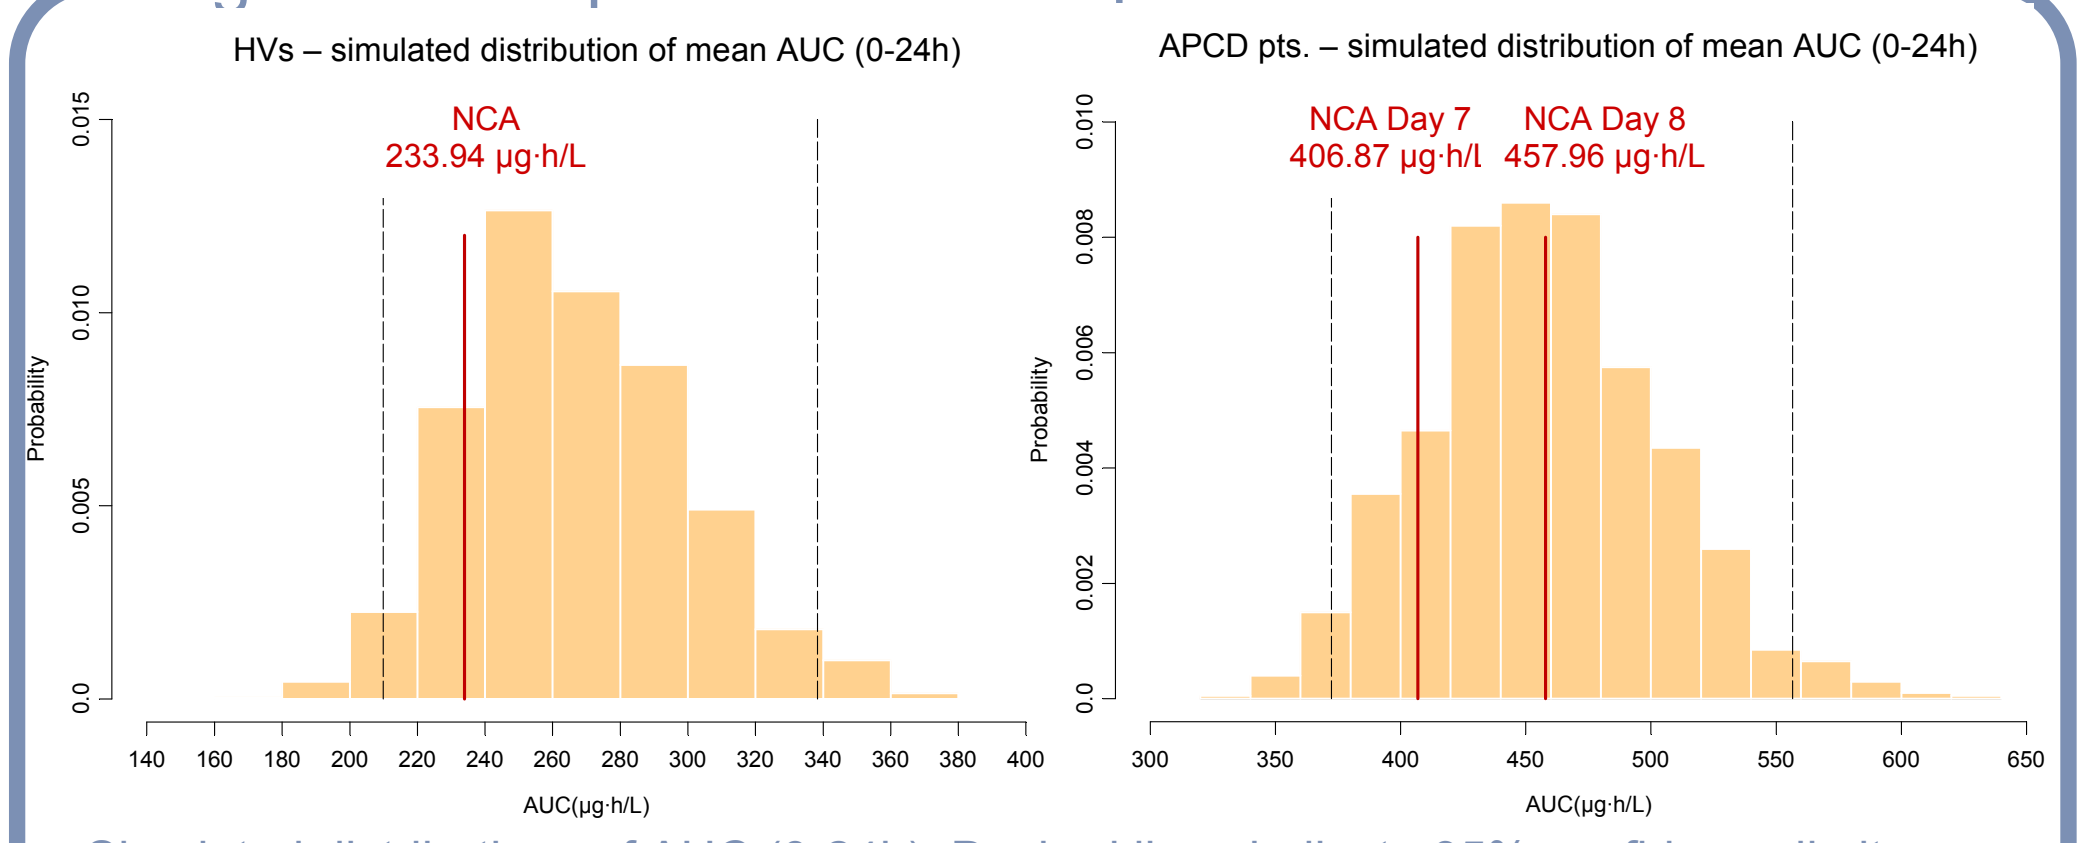
HVs – simulated distribution of mean AUC (0-24h)
NCA
233.94 µg·h/L
Probability
AUC(µg·h/L)
APCD pts. – simulated distribution of mean AUC (0-24h)
NCA Day 7
406.87 µg·h/L
NCA Day 8
457.96 µg·h/L
Probability
AUC(µg·h/L)

Simulated distributions of AUC (0-24h). Dashed lines indicate 95% confidence limits. NCA: Results from the non-compartmental analysis.

Figure 4. Population mean hGH profiles

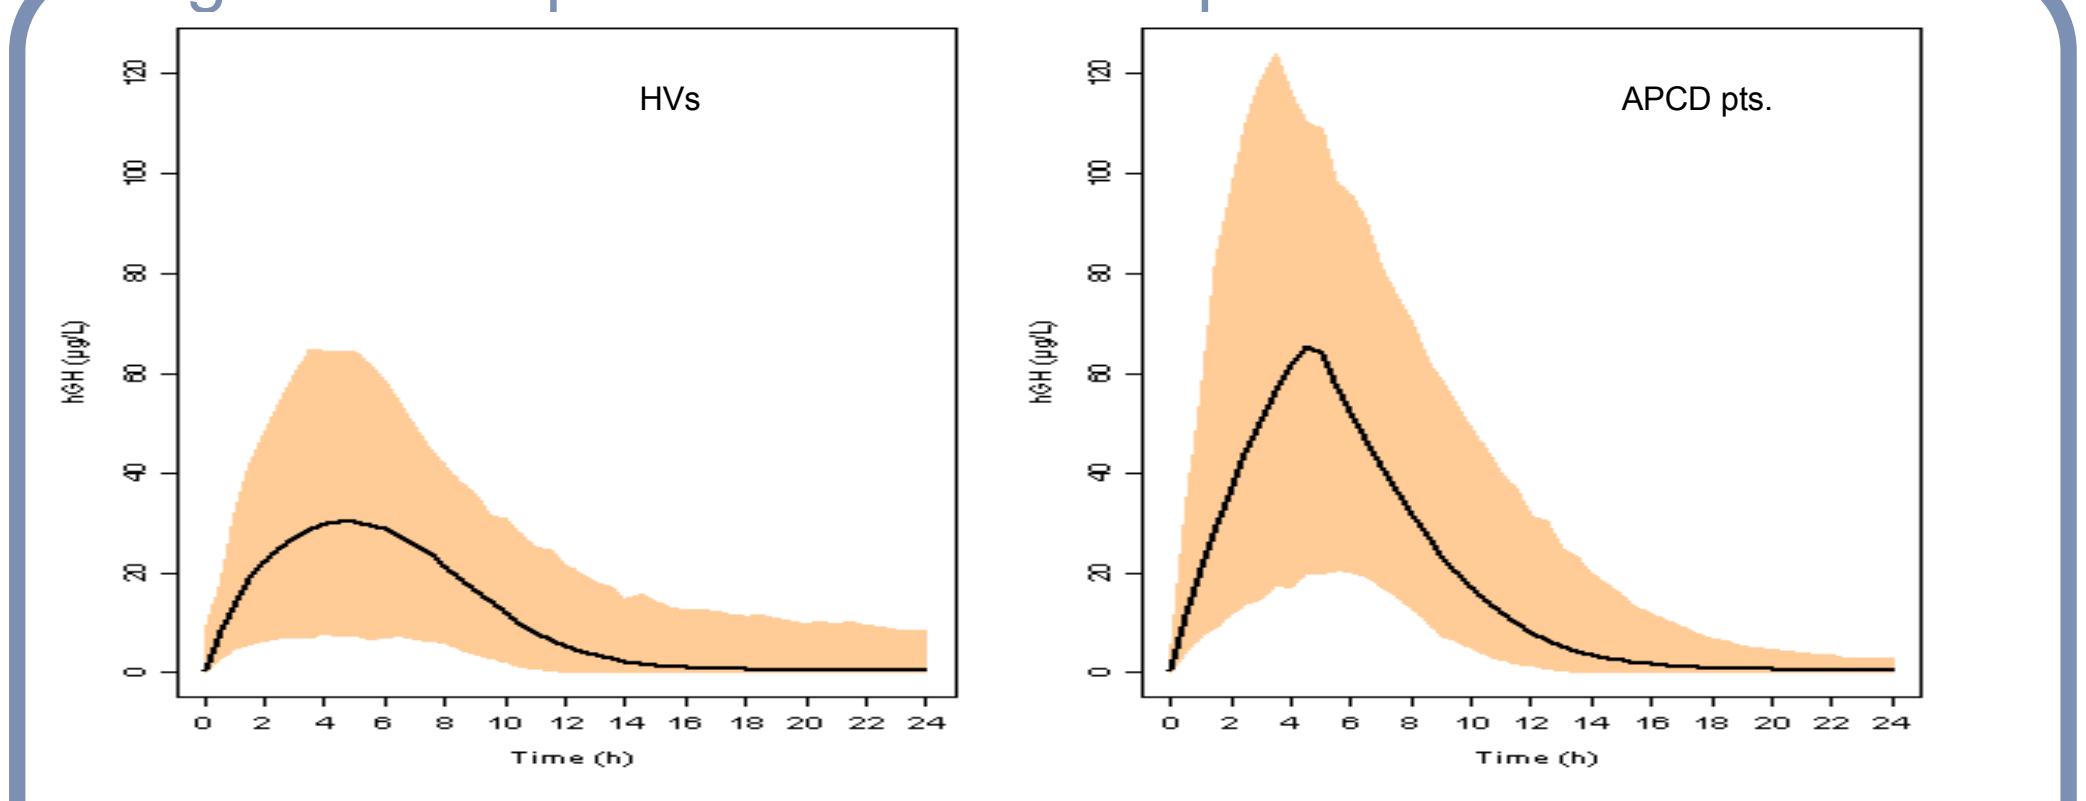
HVs
APCD pts.
hGH (µg/L)
Time (h)

Population mean hGH profiles for HVs (left) and APCD patients (right) on day 7, with 95 % confidence limits. Dose: 50 µg/kg/day for 7 days Based on 1000 simulated subjects in each group.

Table 1. Final model parameters

Parameter	Population Mean	Uncertainty of estimate (% CV of pop mean)	Variability (% CV between subjects)
KMA – HVs (µg/kg)	18.8	44.1	179†
KMA – APCD pts. (µg/kg)	1.06	68.0	179†
VMA (µg/kg/h)	11.3	9.82	46.3†
KM (µg/kg)	18.9	12.6	27.2
VM – HVs (µg/kg/h)	13.0	11.5	-
VM – APCD pts. (µg/kg/h)	9.37	8.38	-
V/f (L/kg)	0.450	5.93	27.5
BASE (µg/L)	0.520	11.3	53.4
q_1 – residual error model†	0.129	7.26	-
q_2 – residual error model†	0.945	13.5	-

BASE: Baseline level of drug , VMA, VM: Maximum absorption, elimination rate, KMA, KM: Amount corres-ponding to half-maximum absorption, elimination rate, %CV of mean: Standard error for mean estimate*100/mean estimate, %CV between subjects: Standard error of individual estimates*100/mean estimate´. †KMA and VMA were correlated with a correlation coefficient of 0.647, †† Estimated parameters in the residual error model $Y_{observed} = f + \sqrt{\theta_1^2 f^2 + \theta_2^2} \cdot \varepsilon$ where *f* is the model predicted, individual *Y* and $\varepsilon \sim N(0,1)$.

Conclusions

- A popPK model of hGH was developed for APCD pts. and HVs.
- Absorption and elimination were found to be different in the HVs and APCD pts.
- Simplified posterior predictive check of the model showed an acceptable performance for simulation purposes.